FPN: 868P

Phase 2 trial of ozuriftamab vedotin (BA3021), a conditionally active biologic (CAB)-ROR2-ADC, in patients with recurrent or metastatic squamous cell carcinoma of the head and neck

Winston Wong,[1] Douglas Adkins,[2] Jamal Misleh,[3] Jochen Lorch,[4] Jaspreet Grewal,[5] Jeffery Russell,[6] Jeremy Cetnar,[7] Alan Ho,[1] Kyechin Chen,[8] Kartik Aysola,[8] Jacob Thomas[9]

[1]Memorial Sloan-Kettering Cancer Center, New York, NY, USA; [2]Washington University School of Medicine, St. Louis, MO, USA; [3]Christiana Hospital, Newark, DE, USA; [4]Northwestern University, Chicago, IL, USA; [5]Norton Cancer Institute, Louisville, KY, USA; [6]Huntsman Cancer Institute, Salt Lake City, UT, USA; [7]Oregon Health & Science University, Portland, OR, USA; [8]BioAtla, Inc., San Diego, CA, USA; [9]USC Norris Comprehensive Cancer Center, Los Angeles, CA, USA

Background

- Despite advancements in therapy, up to 95% of patients with recurrent or metastatic squamous cell carcinoma of the head and neck (R/M SCCHN) ultimately die from the disease.[1]
- ROR2 is overexpressed in many solid tumors, including head and neck, lung, breast, and melanoma, and is associated with resistance to therapies.[2-5]
- Ozuriftamab vedotin (BA3021) is a CAB ROR2 antibody–drug conjugate (CAB-ROR2-ADC) employing an MMAE payload and is being developed for patients with advanced solid tumors.
 - CABs are designed to reduce off-tumor toxicity; this novel mechanism avoids tissue-mediated drug disposition and improves pharmacokinetics.[6]
 - Not impacted by resistance to small molecules or monoclonal antibodies to other targets.
 - Ozuriftamab vedotin conditionally and reversibly binds to the novel ROR2 target under the low-pH conditions of the tumor microenvironment, thus sparing normal tissues **(Figures 1 and 2)**.
 - RP2D (1.8 mg/kg Q2W) was determined from the phase 1 trial (NCT03504488).
 - Ozuriftamab vedotin was recently granted FDA Fast Track Designation for treatment of patients with R/M SCCHN.[7]

Figure 1. Selective CAB binding in the acidic tumor microenvironment

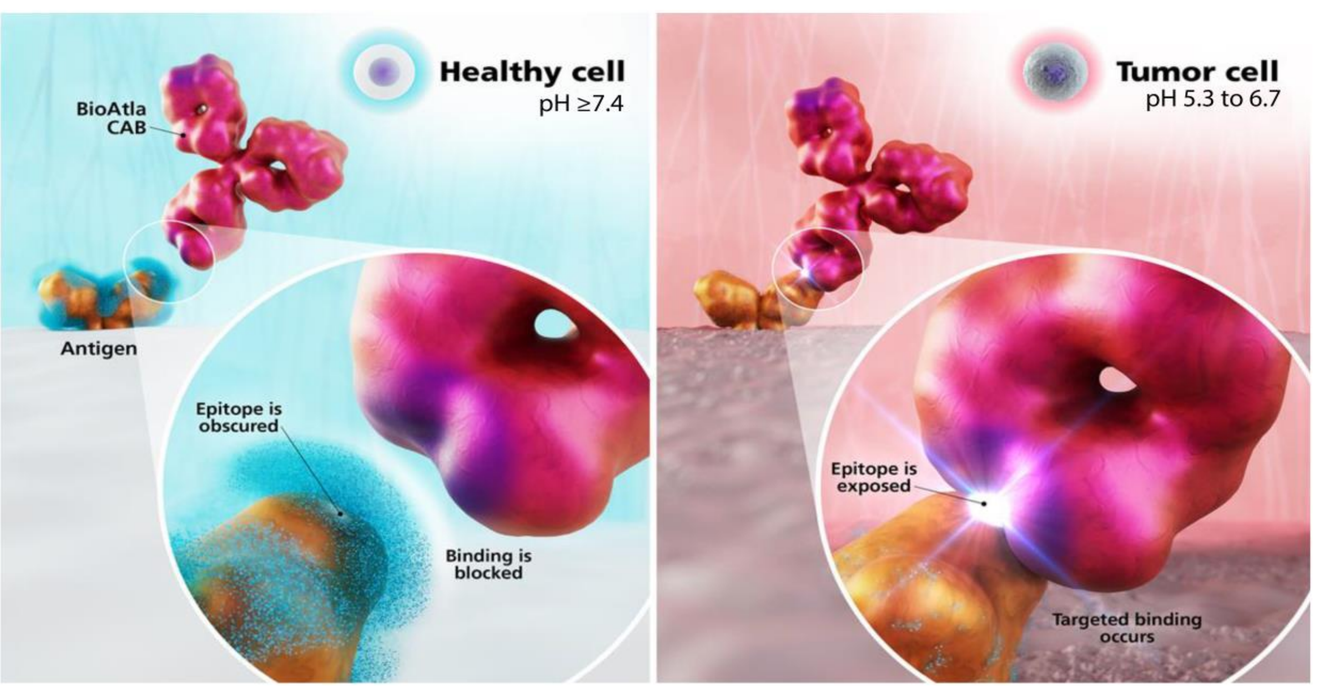
Healthy cell
pH ≥7.4
Tumor cell
pH 5.3 to 6.7
BioAtla CAB
Antigen
Epitope is obscured
Binding is blocked
Epitope is exposed
Targeted binding occurs

The acidity of the tumor cell surface unveils binding sites on targets that are normally shielded in a normal physiologic pH environment. CABs bind to these unveiled sites on cancer cells that are otherwise shielded on healthy cells. CABs are not masked or caged and do not require enzymatic cleavage for activation.

Figure 2. pH-dependent binding of ozuriftamab vedotin to human ROR2

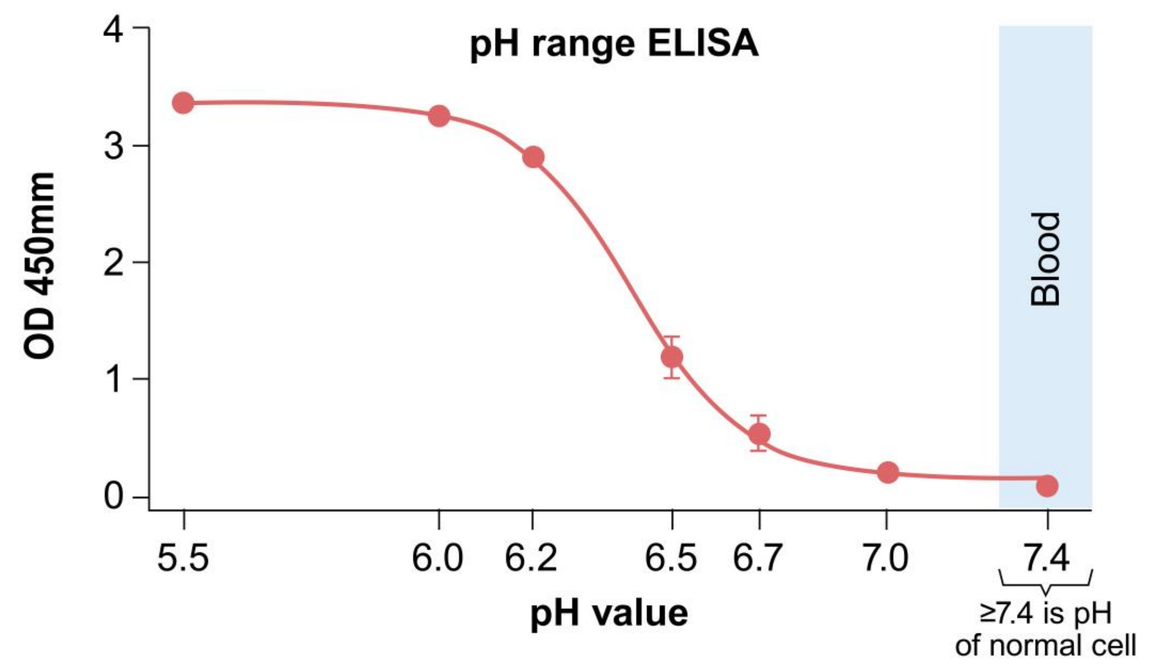
pH range ELISA
OD 450mm
pH value
Blood
5.5
6.0
6.2
6.5
6.7
7.0
7.4
≥7.4 is pH of normal cell

Binding of ozuriftamab vedotin to human ROR2 at different pH conditions was determined by ELISA. CABs preferentially bind at tumor pHs below 6.7, while not binding at the pH of healthy cells (≥7.4).

Methods

Figure 3. Multicenter, open-label, phase 2 study (NCT05271604)

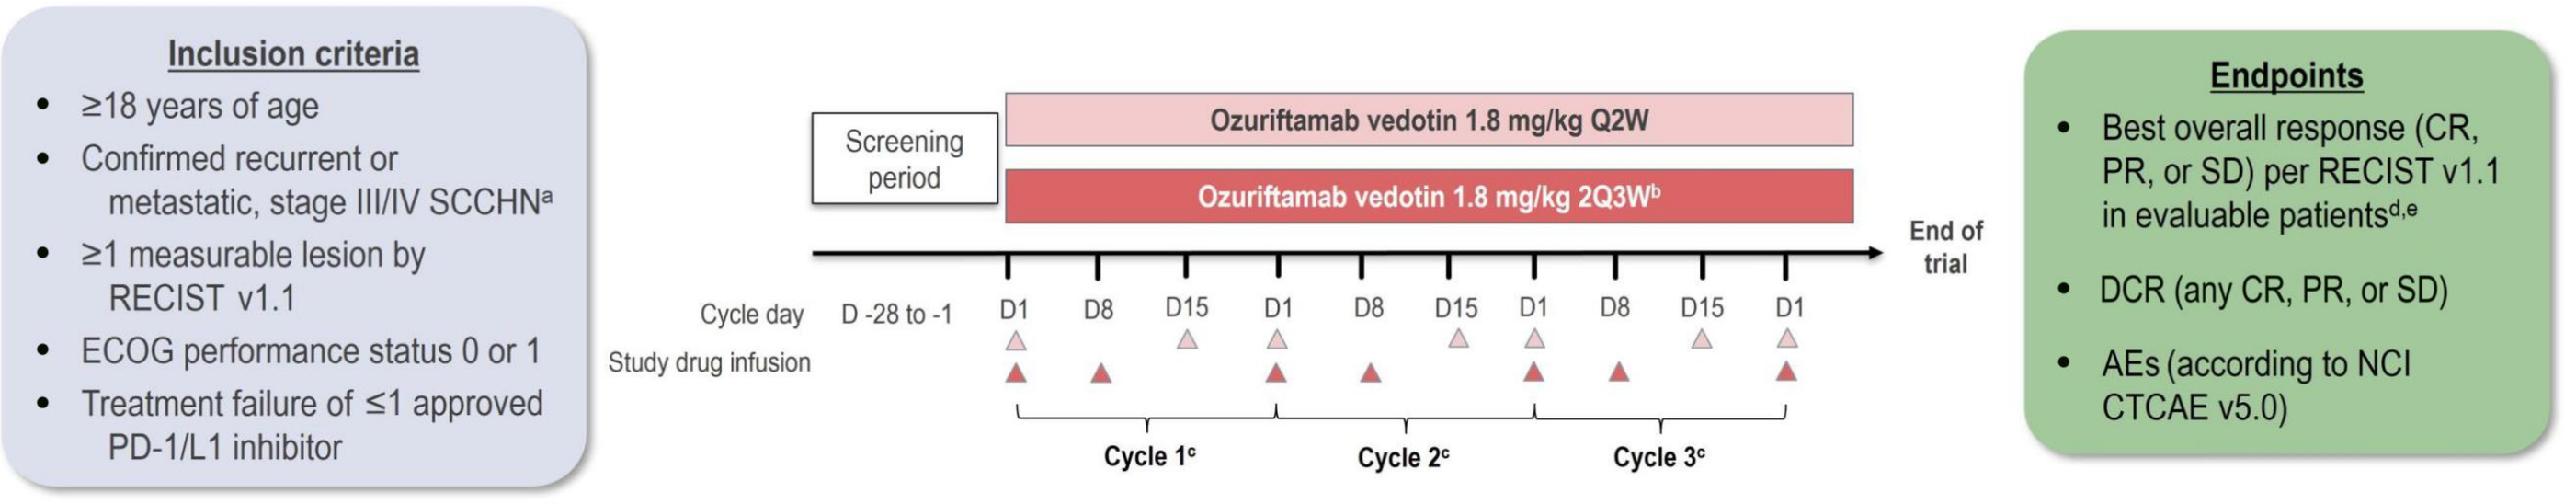
Inclusion criteria
≥18 years of age
Confirmed recurrent or metastatic, stage III/IV SCCHN[a]
≥1 measurable lesion by RECIST v1.1
ECOG performance status 0 or 1
Treatment failure of ≤1 approved PD-1/L1 inhibitor
Screening period
Ozuriftamab vedotin 1.8 mg/kg Q2W
Ozuriftamab vedotin 1.8 mg/kg 2Q3W[b]
End of trial
Cycle day
D -28 to -1
D1 D8 D15 D1 D8 D15 D1 D8 D15 D1
Study drug infusion
Cycle 1[c]
Cycle 2[c]
Cycle 3[c]
Endpoints
Best overall response (CR, PR, or SD) per RECIST v1.1 in evaluable patients[d,e]
DCR (any CR, PR, or SD)
AEs (according to NCI CTCAE v5.0)

[a]Not amenable to local therapy with curative intent (surgery or radiation therapy with or without chemotherapy).
[b]Patients in this dosing regimen were enrolled once Q2W enrollment was complete.
[c]Treatment was given in 28-day cycles in the Q2W cohort and in 21-day cycles in the 2Q3W cohort.
[d]Evaluable patients were defined as those who had ≥1 tumor scan after receiving the study drug.
[e]Tumor assessment by CT or MRI every 6 weeks from C1D1 until 12 weeks, then every 8 weeks up to 1 year, then every 12 weeks thereafter.

Results

Patient disposition

- As of the data cutoff date of May 31, 2024, 32 patients received ozuriftamab vedotin either Q2W (n=12) or 2Q3W (n=20) for a median of 84 days.
- Patients had received a median of 3 prior lines of therapy and had experienced failure of anti–PD-1 therapy **(Table 1)**.

Efficacy

- Overall (31 evaluable patients): 10 responses (32%; 1 CR and 9 PR) and 14 SD **(Table 2 and Figure 4)**.
- Best overall responses were observed regardless of HPV status and ROR2 expression.
 - Q2W (n=12): 1 CR, 2 PR, and 5 SD **(Figure 5)**.
 - 2Q3W (n=19): 7 PR and 9 SD.
 - Median DOR for all confirmed responders has not yet been reached (>3.6 months; 95% CI, 0.4–NE).

Safety

- Most AEs were low grade **(Table 3 and Figure 6)**; fatigue (59%), anemia (34%), and nausea (34%) were the most frequent AEs.
- Six patients (19%) had grade 3 TRAEs (nausea, diarrhea, decreased lymphocyte count, decreased neutrophil count, peripheral neuropathy, elevated liver enzymes, hyperglycemia, soft tissue infection, febrile neutropenia, asthenia, and dysphagia).
 - One patient (3%) experienced a TRAE of grade 4 hyponatremia.
 - No grade 5 TRAEs were observed.
- Most frequent grade 3-4 TEAEs were hyponatremia (13%), decreased lymphocyte count (13%), anemia (9%), and hypoxia (9%) **(Figure 6)**.
- Two patients experienced related AEs leading to study drug discontinuation (peripheral neuropathy in Q2W and 2Q3W).

Results (continued)

Table 1. Patient demographics and clinical characteristics

Ozuriftamab vedotin 1.8 mg/kg	Q2W (n=12)	2Q3W (n=20)	Total (N=32)
Age, mean (SD), y	62 (11)	66 (7)	64 (9)
Sex, n (%)			
Male	11 (92)	18 (90)	29 (91)
Female	1 (8)	2 (10)	3 (9)
ECOG performance, n (%)			
0	5 (42)	8 (40)	13 (41)
1	7 (58)	12 (60)	19 (59)
Location of primary disease, n (%)			
Oropharynx	11 (92)	14 (70)	25 (78)
Oral cavity	1 (8)	3 (15)	4 (13)
Hypopharynx	0	1 (5)	1 (3)
Larynx	2 (17)	3 (15)	5 (16)
Other location	2 (17)	5 (25)	7 (22)
Number of prior lines of therapy, median	3	3	3
Prior anti–PD-1 exposure, n (%)	12 (100)	20 (100)	32 (100)
Prior platinum-based chemotherapy exposure, n (%)	11 (92)	20 (100)	31 (97)
HPV status,[a] n (%)			
Positive	4 (33)	5 (25)	9 (28)
Negative	1 (8)	3 (15)	4 (13)
Missing/not reported	7 (58)	12 (60)	19 (59)

[a]HPV status was determined using p16 immunohistochemistry.

Table 2. Best overall response

Ozuriftamab vedotin 1.8 mg/kg	Q2W (n=12)	2Q3W (n=19)	Total (N=31[a])
Any response[b] (confirmed and unconfirmed CR or PR)			
Responses, n (%)	3 (25)	7 (37)	10 (32)
95% CI	6, 57	16, 62	17, 51
Best overall response[c], n (%)			
Confirmed CR	1 (8)	0	1 (3)
PR with/without confirmation	1 (8) / 1 (8)	4 (21) / 3 (16)	5 (16) / 4 (13)
SD	5 (42)	9 (47)	14 (45)
Disease control rate[b,d]			
DCR, n (%)	8 (67)	16 (84)	24 (77)
95% CI	35, 90	60, 97	59, 90

[a]1 patient withdrew consent prior to tumor assessment.
[b]Two-sided 95% CIs were calculated using the exact probability method based on the binomial distribution.
[c]If the best overall response of a patient was an unconfirmed CR and a confirmed PR, then the patient was summarized under PR with confirmation.
[d]Disease control was defined as any CR, PR, or SD.

Figure 4. Percentage change from baseline in sum of target lesions

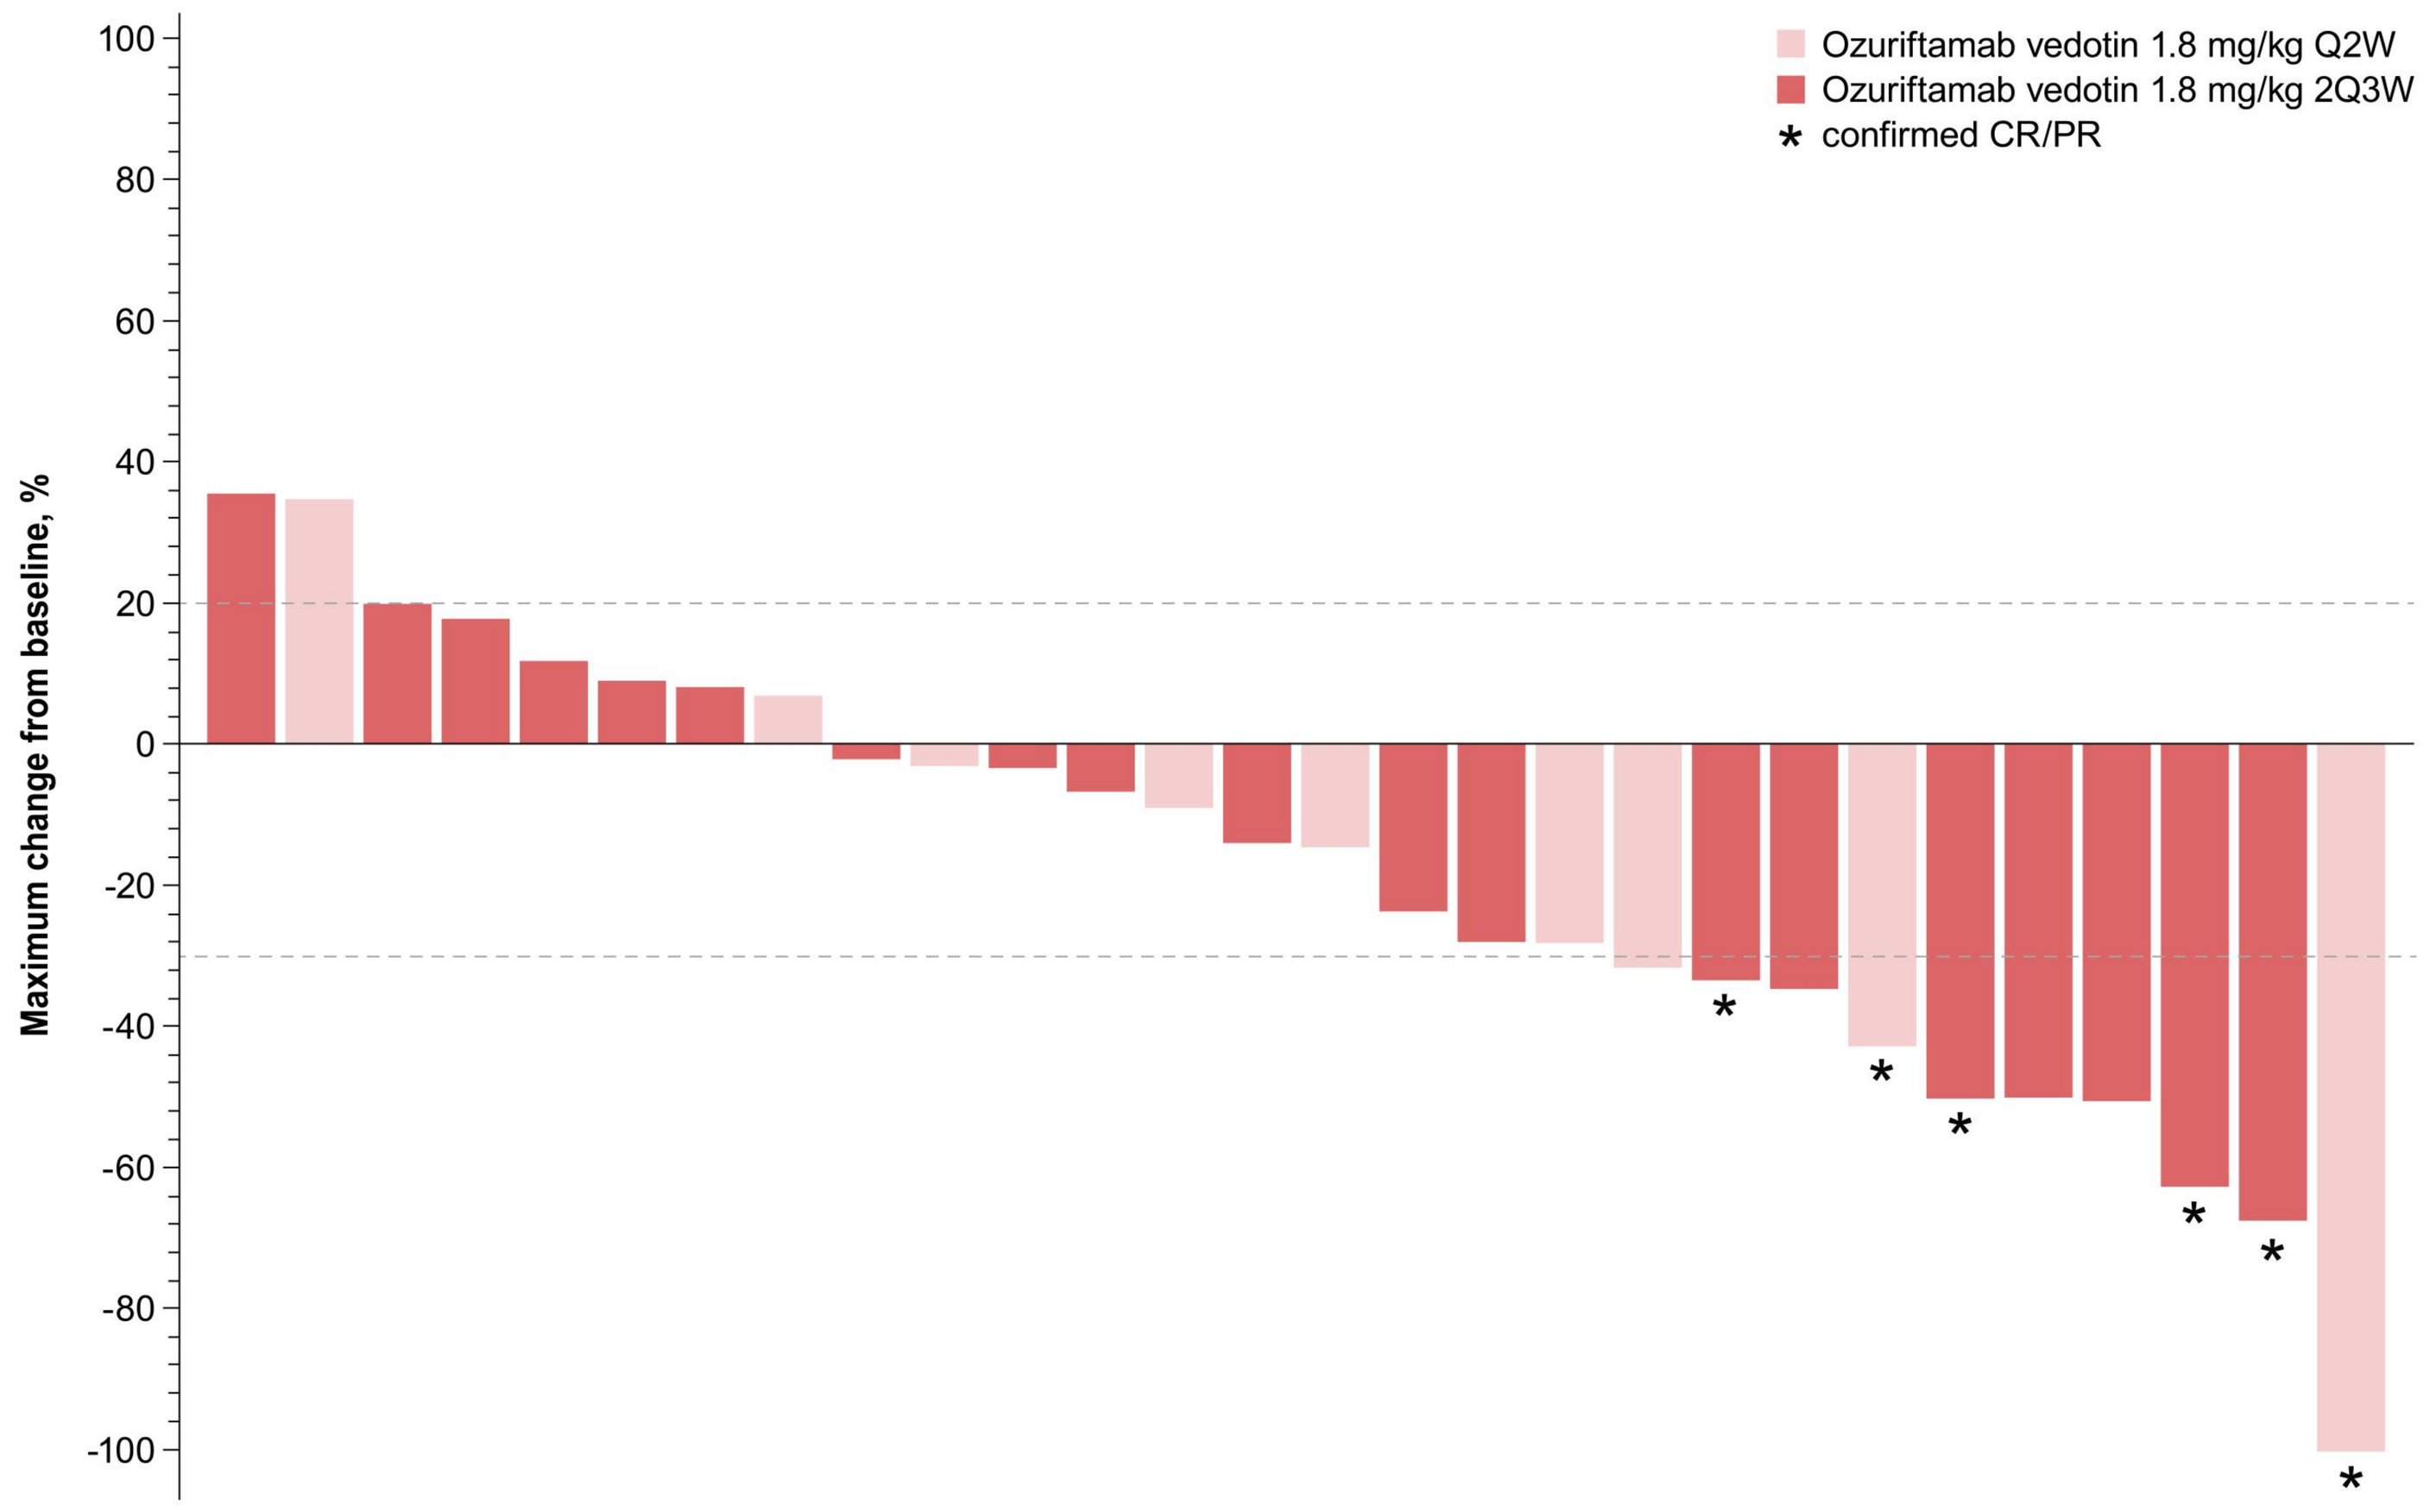
Ozuriftamab vedotin 1.8 mg/kg Q2W
Ozuriftamab vedotin 1.8 mg/kg 2Q3W
* confirmed CR/PR
Maximum change from baseline, %
100
80
60
40
20
0
-20
-40
-60
-80
-100

Table 3. Summary of AEs

Number of patients with any AE, n (%)	Q2W (n=12)	2Q3W (n=20)	Total (N=32)
AE	11 (92)	20 (100)	31 (97)
Related	8 (67)	19 (95)	27 (84)
≥Grade 3 AE[a]	8 (67)	14 (70)	22 (69)
Related grade 3	1 (8)	5 (25)	6 (19)
Related grade 4	0	1 (3)	1 (3)
Serious AE	8 (67)	9 (45)	17 (53)
Related	1 (8)	3 (15)	4 (13)
AE leading to treatment discontinuation	2 (17)	1 (5)	3 (9)
Related	1 (8)	1 (5)	2 (6)
AE leading to death	0	1 (5)	1 (3)
Related	0	0	0

[a]No grade 5 related AEs observed.

Conclusions

- **ROR2 is a receptor tyrosine kinase involved in Wnt signal transduction that contributes to treatment resistance in advanced cancer.[5]**
- **Treatment with ozuriftamab vedotin achieved durable responses and promising tumor control among a patient population with a median of 3 prior lines of therapy.**
- **Ozuriftamab vedotin was remarkably well tolerated in the context of other available therapies.**
- **Further evaluation of ozuriftamab vedotin monotherapy versus investigator's choice in platinum/PD-1 treatment-refractory patients is planned.**

Figure 5. Confirmed CR in a 78-year-old male with a history of p16+ base of tongue squamous cell cancer diagnosed in 2018 s/p surgery and adjuvant radiation with multiple tumor recurrences and progression after pembrolizumab (initiated September 2022)

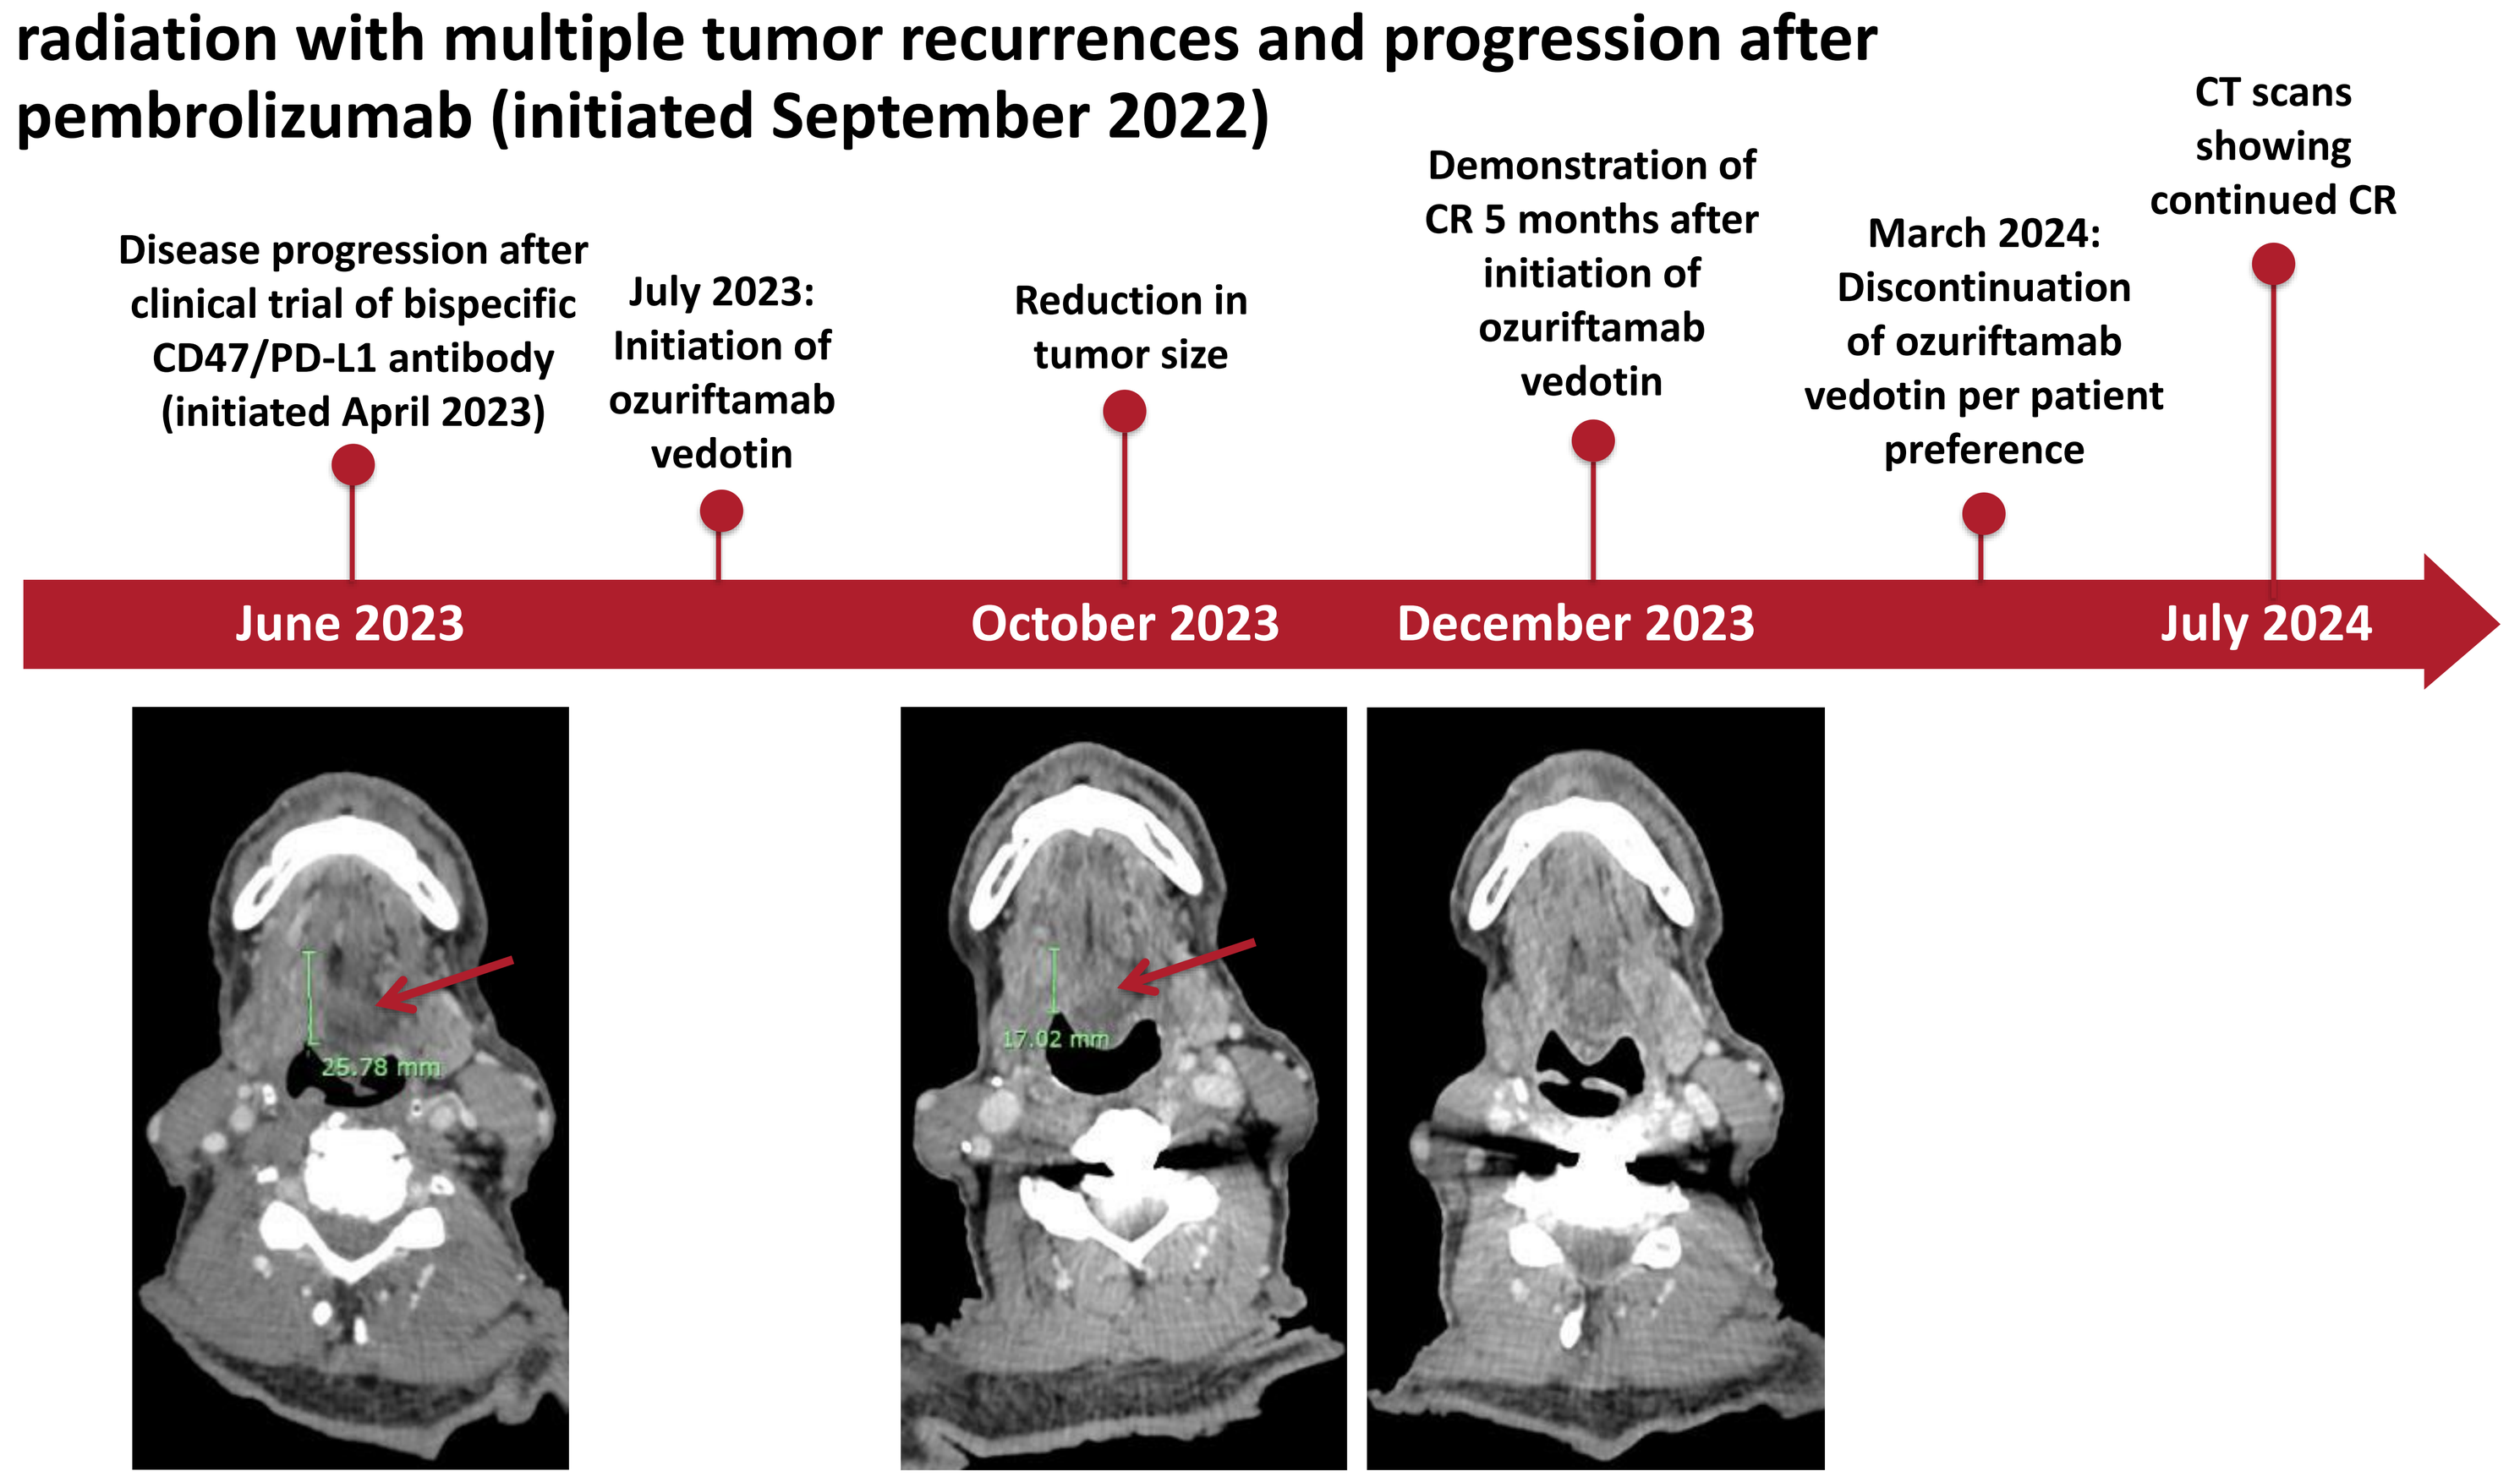
Disease progression after clinical trial of bispecific CD47/PD-L1 antibody (initiated April 2023)
July 2023: Initiation of ozuriftamab vedotin
Reduction in tumor size
Demonstration of CR 5 months after initiation of ozuriftamab vedotin
March 2024: Discontinuation of ozuriftamab vedotin per patient preference
CT scans showing continued CR
June 2023
October 2023
December 2023
July 2024
25.78 mm
17.02 mm

Figure 6. Most frequent AEs of any grade (>16% of patients)

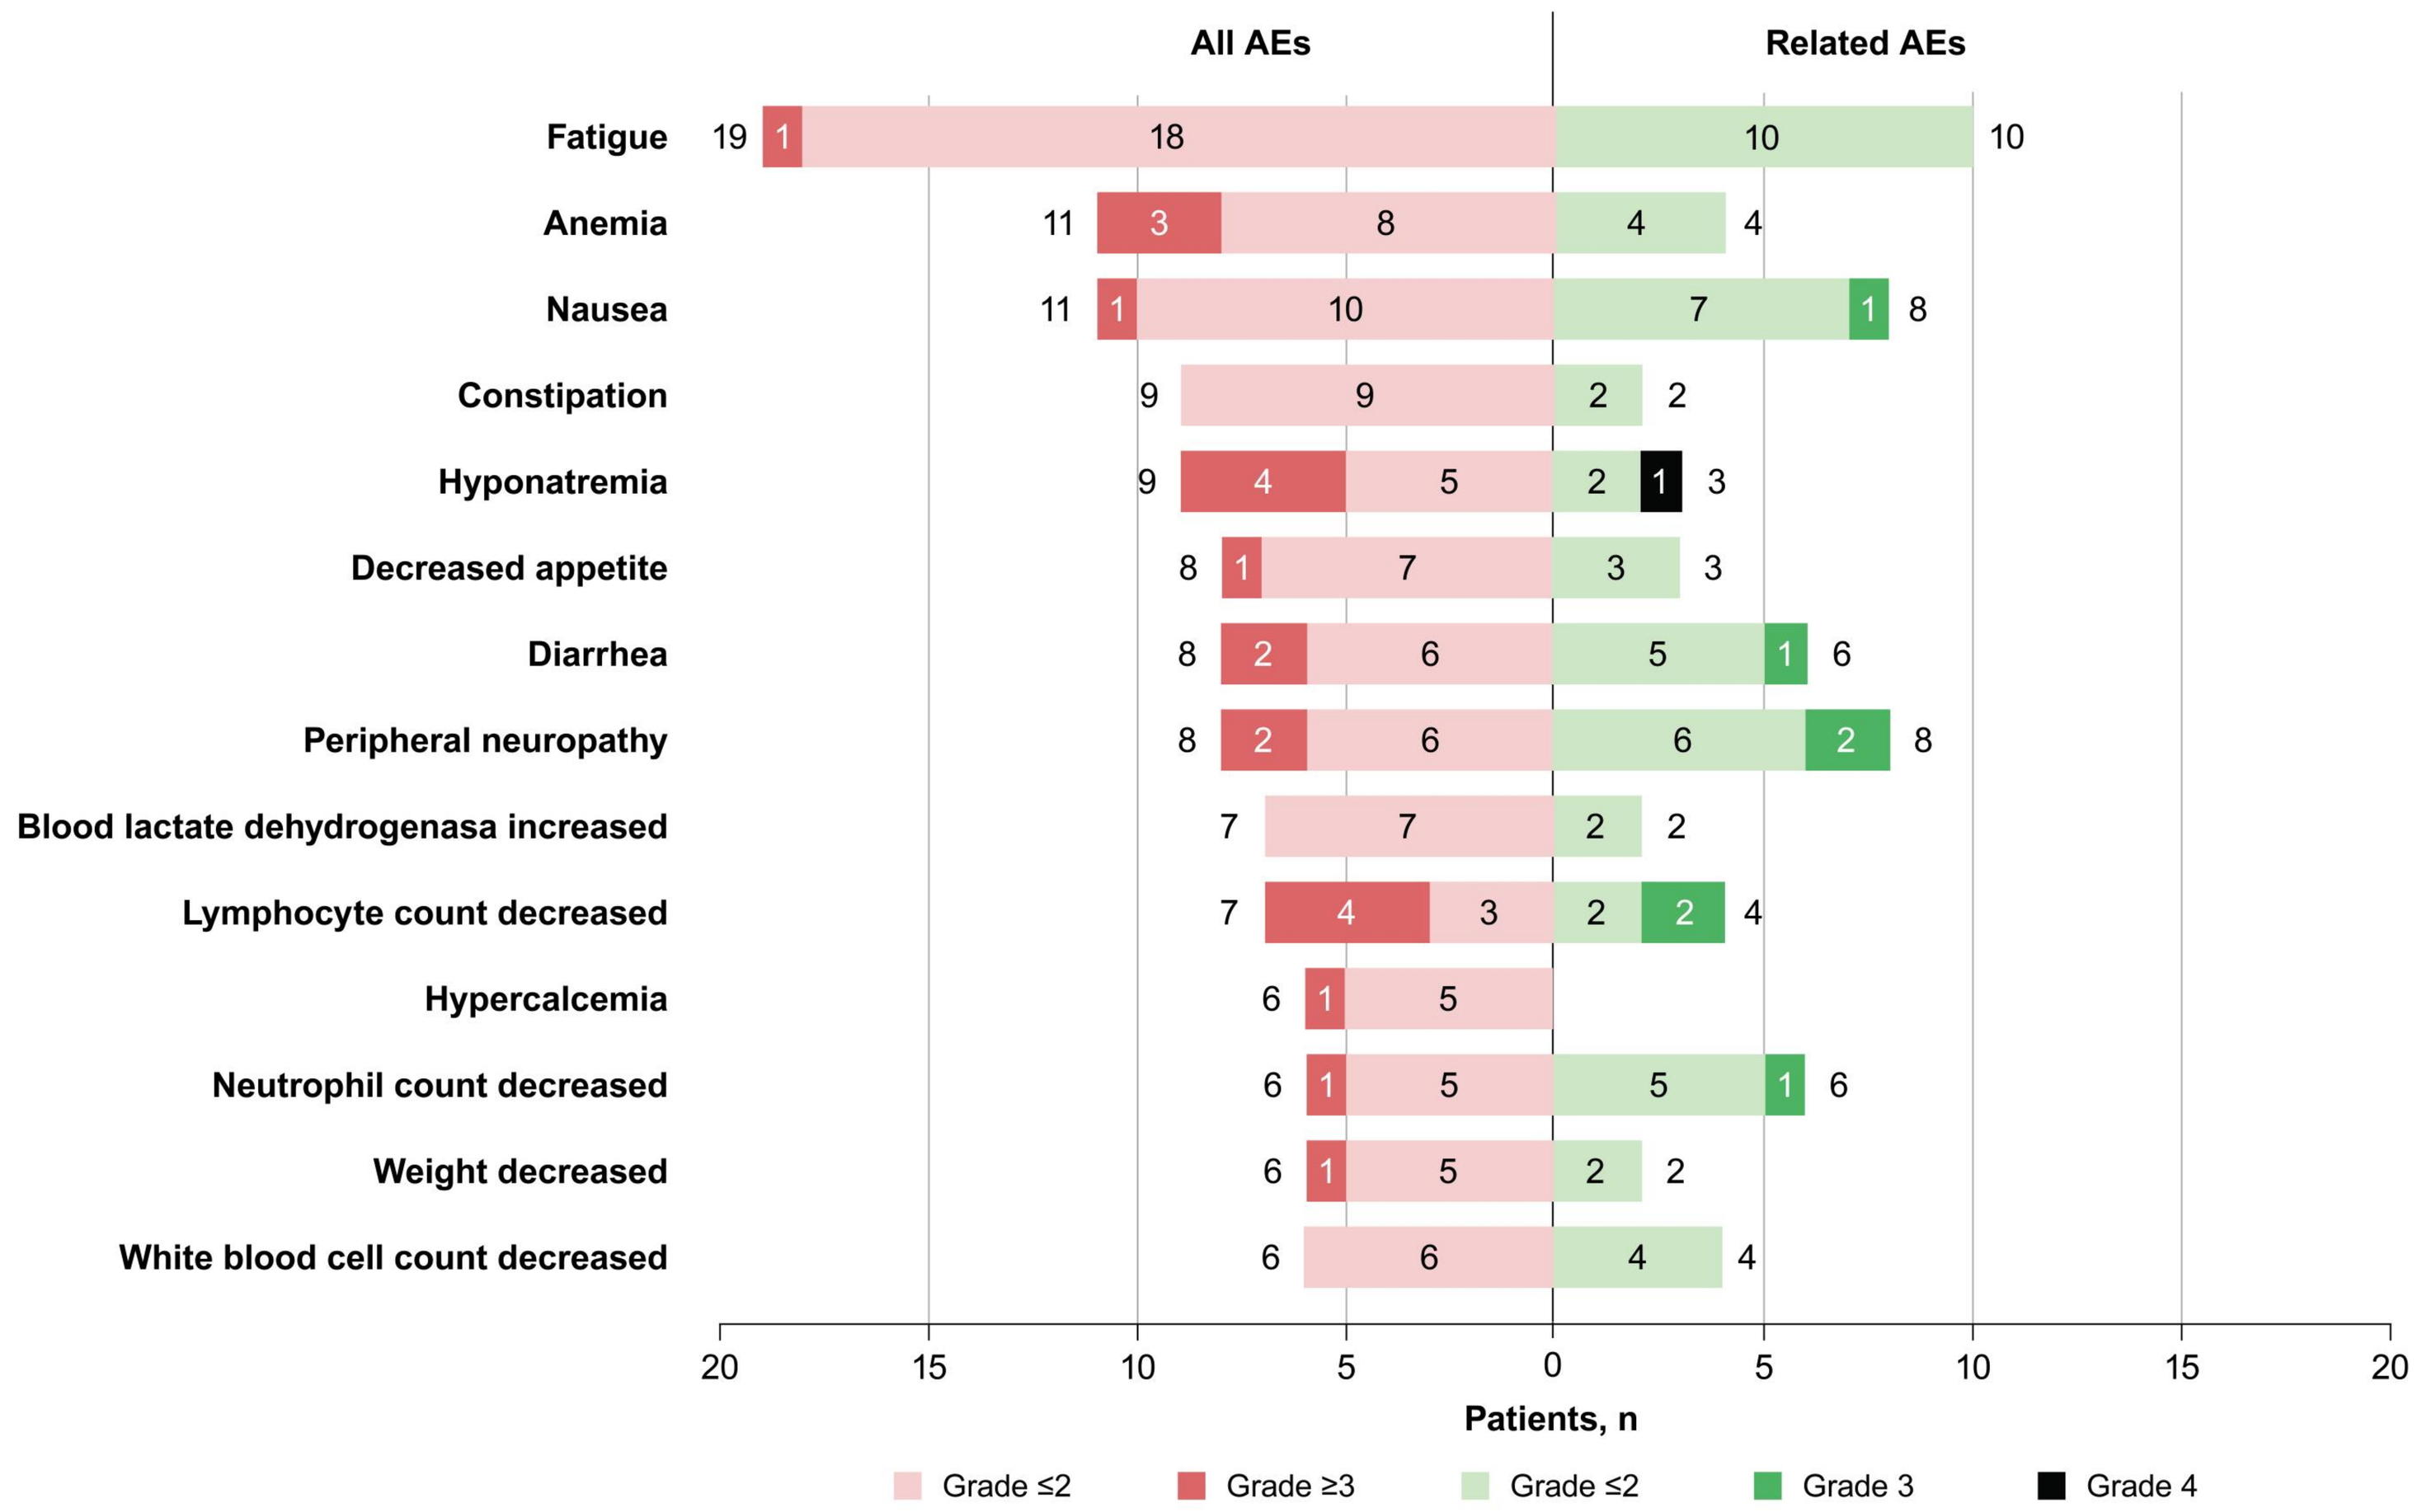
All AEs
Related AEs
Fatigue
Anemia
Nausea
Constipation
Hyponatremia
Decreased appetite
Diarrhea
Peripheral neuropathy
Blood lactate dehydrogenasa increased
Lymphocyte count decreased
Hypercalcemia
Neutrophil count decreased
Weight decreased
White blood cell count decreased
Patients, n
Grade ≤2
Grade ≥3
Grade ≤2
Grade 3
Grade 4

References

1. Chow LQM. *N Engl J Med*. 2020;382(1):60-72.
2. Debebe Z, et al. *Pharmacol Ther*. 2015;150:143-148.
3. Henry C, et al. *J Cancer Res Clin Oncol*. 2015;141(2):243-254.
4. Lu C, et al. *Oncotarget*. 2015;6(28):24912-24921.
5. Castro MV, et al. *Crit Rev Oncol Hematol*. 2022;170:103595.
6. Chang HW, et al. *Proc Natl Acad Sci*. 2021;118(9):e2020606118.
7. BioAtla granted FDA fast track designation for ozuriftamab vedotin (CAB-ROR2-ADC) for treatment of patients with recurrent or metastatic squamous cell carcinoma of the head and neck. BioAtla, Inc. July 23, 2024. Accessed September 6, 2024.

Abbreviations

2Q3W, days 1 and 8 every 3 weeks; AE, adverse event; C, cycle; CAB, conditionally active biologic; CR, complete response; CT, computed tomography; CTCAE, Common Terminology Criteria for Adverse Events; D, day; DCR, disease control rate; DOR, duration of response; ECOG, Eastern Cooperative Oncology Group; ELISA, enzyme-linked immunosorbent assay; FDA, US Food and Drug Administration; HPV, human papillomavirus; MMAE, monomethyl auristatin E; MRI, magnetic resonance imaging; NCI, National Cancer Institute; NE, not estimable; NR, not reached; OD, optical density; PD-1, programmed cell death protein 1; PD-L1, programmed cell death ligand 1; PR, partial response; Q2W, days 1 and 15 every 2 weeks; RECIST, Response Evaluation Criteria in Solid Tumors; ROR2, receptor tyrosine kinase orphan receptor 2; RP2D, recommended phase 2 dose; s/p, status post; SD, stable disease; v, version; TEAE, treatment-emergent adverse event; TRAE, treatment-related adverse event.

Acknowledgments and Funding

This study was funded by BioAtla, Inc. Medical writing support was provided by Alec Jacobson, MD, and Vivek Shah, PhD, from and for MedLogix Communications, LLC, respectively, a Citrus Health Group, Inc., company (Chicago, IL), and was funded by BioAtla, Inc.

Disclosures

WW: Research/PI support from BioAtla and Hookipa.

Correspondence: Kartik Aysola, MD (MedicalAffairs@bioatla.com)

Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors.

Presented at European Society for Medical Oncology • Barcelona, Spain • September 13–17, 2024